Strategies for diverse participant recruitment to a preclinical Alzheimer's disease trial: The AHEAD Study

Doris P Molina-Henry, PhD[1,2], Crystal M Glover, PhD[3], Joshua D Grill, PhD[4], Reisa A Sperling, MD[5], Paul Aisen, MD[1,2], Taylor Clanton, MPH[1,2], Theresa Devins, DrPH, MS[6], , Chad J. Swanson, PhD[6] Shobha Dhadda, PhD[6], Michael C Irizarry, MD, MPH[6], Keith A Johnson, MD[5], Dylan Kirn, MPH[5], Lynn D Kramer, MD FAAN[6], Leona K Shum, MPH [1,2], Jin Zhou MD, PhD[6] and Rema Raman, PhD[1,2]

[1]Alzheimer's Therapeutic Research Institute, [2]University of Southern California, San Diego, CA, USA; [3]Rush University Medical Center, Chicago, IL, USA [4]University of California, Irvine; CA, USA [5]Massachusetts General Hospital, Boston, MA, USA ; [6]Eisai Inc., Nutley, NJ, USA

P1-13

BACKGROUND

Recruitment of demographically diverse populations is imperative to the generalizability of results from Alzheimer's disease (AD) clinical trials and to ensure representation of all communities affected by dementia.

- Compared to non-Hispanic Whites, Black/African Americans and Hispanic/Latino(a)s adults are 2 and 1.5 times more likely to develop AD, respectively. These and other demographically diverse communities are greatly affected by AD and yet are underrepresented in trials.
- The Alzheimer's Clinical Trials Consortium (ACTC) is committed to the testing and implementation of focused and innovative approaches that promote the participation of individuals from demographically diverse backgrounds in AD clinical trials.

We provide preliminary data on the impact of earned media interviews on the recruitment of Black/African American adults to the AHEAD Study.

METHOD

- The AHEAD Study (NCT04468659), comprised of two clinical trials -A3 and A45- investigates the efficacy of lecanemab (BAN2401) in slowing biological and cognitive disease progression.
- The AHEAD Study enrolls individuals ages 55-80 who are cognitively unimpaired but who are biomarker eligible with elevated levels of amyloid.
- The AHEAD Study Recruitment and Retention Workgroup designed and implemented a recruitment strategy focused on precision earned media recruitment.
- Communication strategies highlighting the importance of Black/African American participation in AD clinical trials were developed with input from investigators who are experts in disparities in AD.
- Earned media interviews with racially congruent AHEAD spokespeople occurred during Black History Month (February).

RESULTS

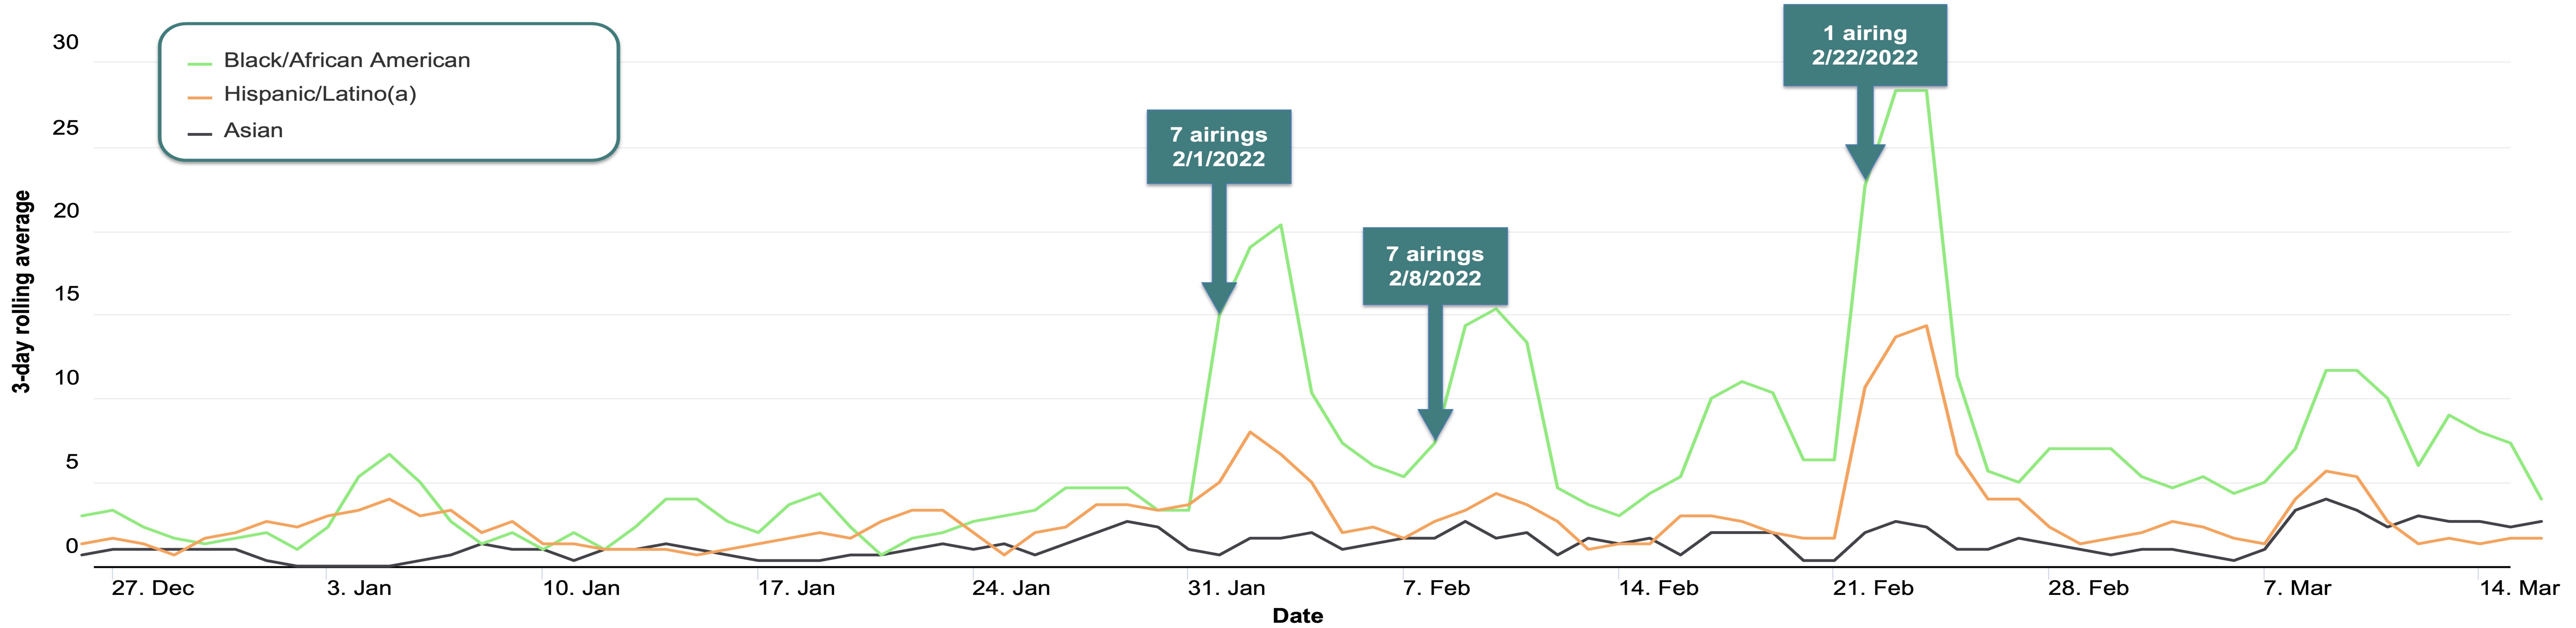

Black/African American
Hispanic/Latino(a)
Asian
7 airings
2/1/2022
7 airings
2/8/2022
1 airing
2/22/2022
3-day rolling average
30
25
20
15
10
5
0
27. Dec
3. Jan
10. Jan
17. Jan
24. Jan
31. Jan
7. Feb
14. Feb
21. Feb
28. Feb
7. Mar
14. Mar
Date

Figure . 3-day rolling average of completed pre-screeners across 3 different racial and ethnic groups during the month of February. Airings focused on the importance of clinical trial participation during Black History Month 2022. Cumulative count of completed pre-screeners throughout the month of February. Notably, an increased average number of daily completed.

Date	Station	Outlet	City
2/1/2022	KTTV (FOX)	TV	Los Angeles, CA
	KCBS (AM)	Radio	San Francisco, CA
	WRGC (FM)	Radio	Sylva, NC
	WGBO (FM)	Radio	Newark NJ/New York City, NY
	WTSP (CBS)	TV	Tampa, FL
	WBFF (FOX)	TV	Baltimore, MD
	WJXT (CW)	TV	Jacksonville, FL
2/8/2022	KTUL-TV (ABC)	TV	Tulsa, OK
	KTXL-TVL (FOX)	TV	Sacramento, CA
	KYWM (FM)	Radio	Philadelphia, PA
	WCSC-TV	TV	Charleston, SC
	WTOP-FM	Radio	Washington, DC
	KCBS-TV (ABC)	TV	Los Angeles, CA
	KPIX-TV (CBS)	TV	San Francisco CA
2/22/2022	KNXV-TV	TV	Phoenix, AZ

Table 2 Dates, media stations, outlets, and cities of airings during Black History Month in February.

- 15 earned media interviews were conducted across national and local radio and TV stations during the month of February (**Table 1**).
- An uptick in pre-screeners completed by individuals who identified as Black/African American was observed following the different airing dates **(Figure)**.
- The number of Black/African Americans who completed the pre-screener tripled in February compared to December and January (**Table 2**).
- Overall number of pre-screeners completed by other underrepresented groups also increased in February compared to December and January.

Month	Black/African American	Hispanic/ Latino(a)	Asian
Dec	97	40	28
Jan	93	70	27
Feb	274	110	42

Table 2 Number of individuals from underrepresented groups that completed the pre-screener across December, January and February.

DISCUSSION

- The AHEAD Study provides an infrastructure through its pre-screen database, www.AHEADstudy.org, to evaluate central recruitment strategies.
- Earned media through trusted outlets, tailored to addressing the importance and value of participation of Black/African American adults in clinical trials is effective for the engagement of prospective participants into preclinical trials.
- Future initiatives will focus on evaluating a similar approach across other demographically diverse groups for their impact and effectiveness in engaging underrepresented populations into the AHEAD Study.

ACKNOWLEDGEMENTS

We would like to acknowledge our research partners Eisai and Biogen. This work was supported by ACTC Grant # U24AG057437