Video imaging of structural dynamics of amyloid β protofibrils

TAKAHIRO WATANABE-NAKAYAMA [a], Mayumi Tsuji [b], Tatsunori Oguchi [b], Kenjiro Ono [c]

[a] Nano Life Science Institute, Kanazawa University, JAPAN

[b] Department of Pharmacology, Division of Medical Pharmacology, School of Medicine, Showa University, JAPAN

[c] Department of Internal Medicine, Division of Neurology, School of Medicine, Showa University, JAPAN

High-speed atomic force microscopy reveals that amyloid β protofibrils show segmented but rigid structure which is stiffer than mature fibrils in liquid, and that lecanemab (BAN2401) binds protofibrils with higher specificity and stronger than 4G8 monoclonal antibody.

Current treatments for Alzheimer's disease (AD) are primarily aimed at mitigating symptoms, while disease-modifying approaches are aimed at halting or attenuating the progression of the disease, such as inhibiting Aβ production and aggregation or promoting Aβ1-42 clearance. Although no disease-modifying drug for AD has been approved despite many long and expensive trials, very recently, the anti-Aβ aggregates selective antibody, aducanumab was approved for the treatment of AD by FDA in US using the accelerated approval pathway. In addition, it was reported that lecanemab, an antibody developed for early AD with a unique target binding profile selective for Aβ protofibrils (PFs), significantly slowed cognitive decline with a concomitant reduction in amyloid plaques [1] and reduced cell toxicity (Fig.1). The Aβ aggregates are dynamic, which makes it difficult to extract and analyze structural dynamics in a stable state. Here, we elucidated the motion dynamics of protofibrils and its molecular interaction with lecanemab using high-speed atomic force microscopy (HS-AFM). HS–AFM allows video–imaging of structural dynamics of individual amyloid aggregates in liquid [2-4].

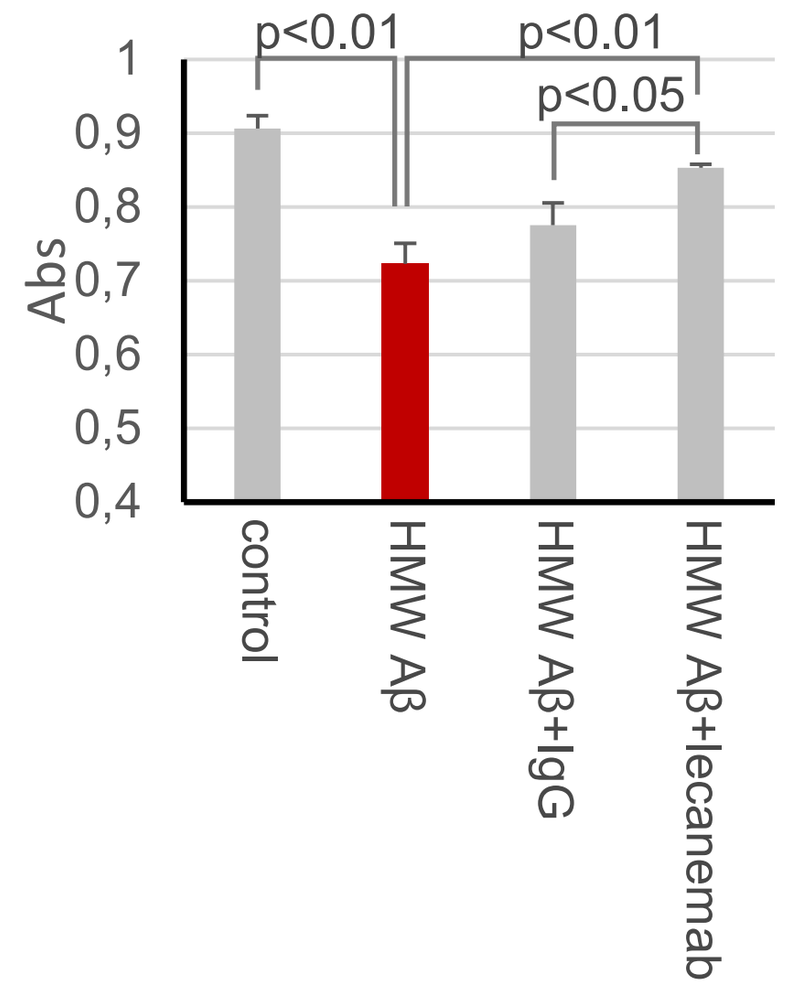

p<0.01
p<0.01
p<0.05
Abs
1
0,9
0,8
0,7
0,6
0,5
0,4
control
HMW Aβ
HMW Aβ+IgG
HMW Aβ+lecanemab

Figure 1. Viability of SH-SY5Y neuroblastoma cells following high molecular weight Aβ oligomers (HMW) exposure and IgG or lecanemab. Cell viability was evaluated by 3-(4,5-Dimethylthiazoyl-2-yl)-2,5-diphenyltetrazolium bromide viable cell (MTT) assay. HMW Aβ-inducing cell toxicity [5] was reduced by the removal of toxic oligomers interacted with lecanemab.

Preparation of Aβ1-42 protofibrils (PFs) and mature fibrils (MFs)

We collected PFs and MFs from incubation of synthetic Aβ1-42 peptides (Fig. 2)

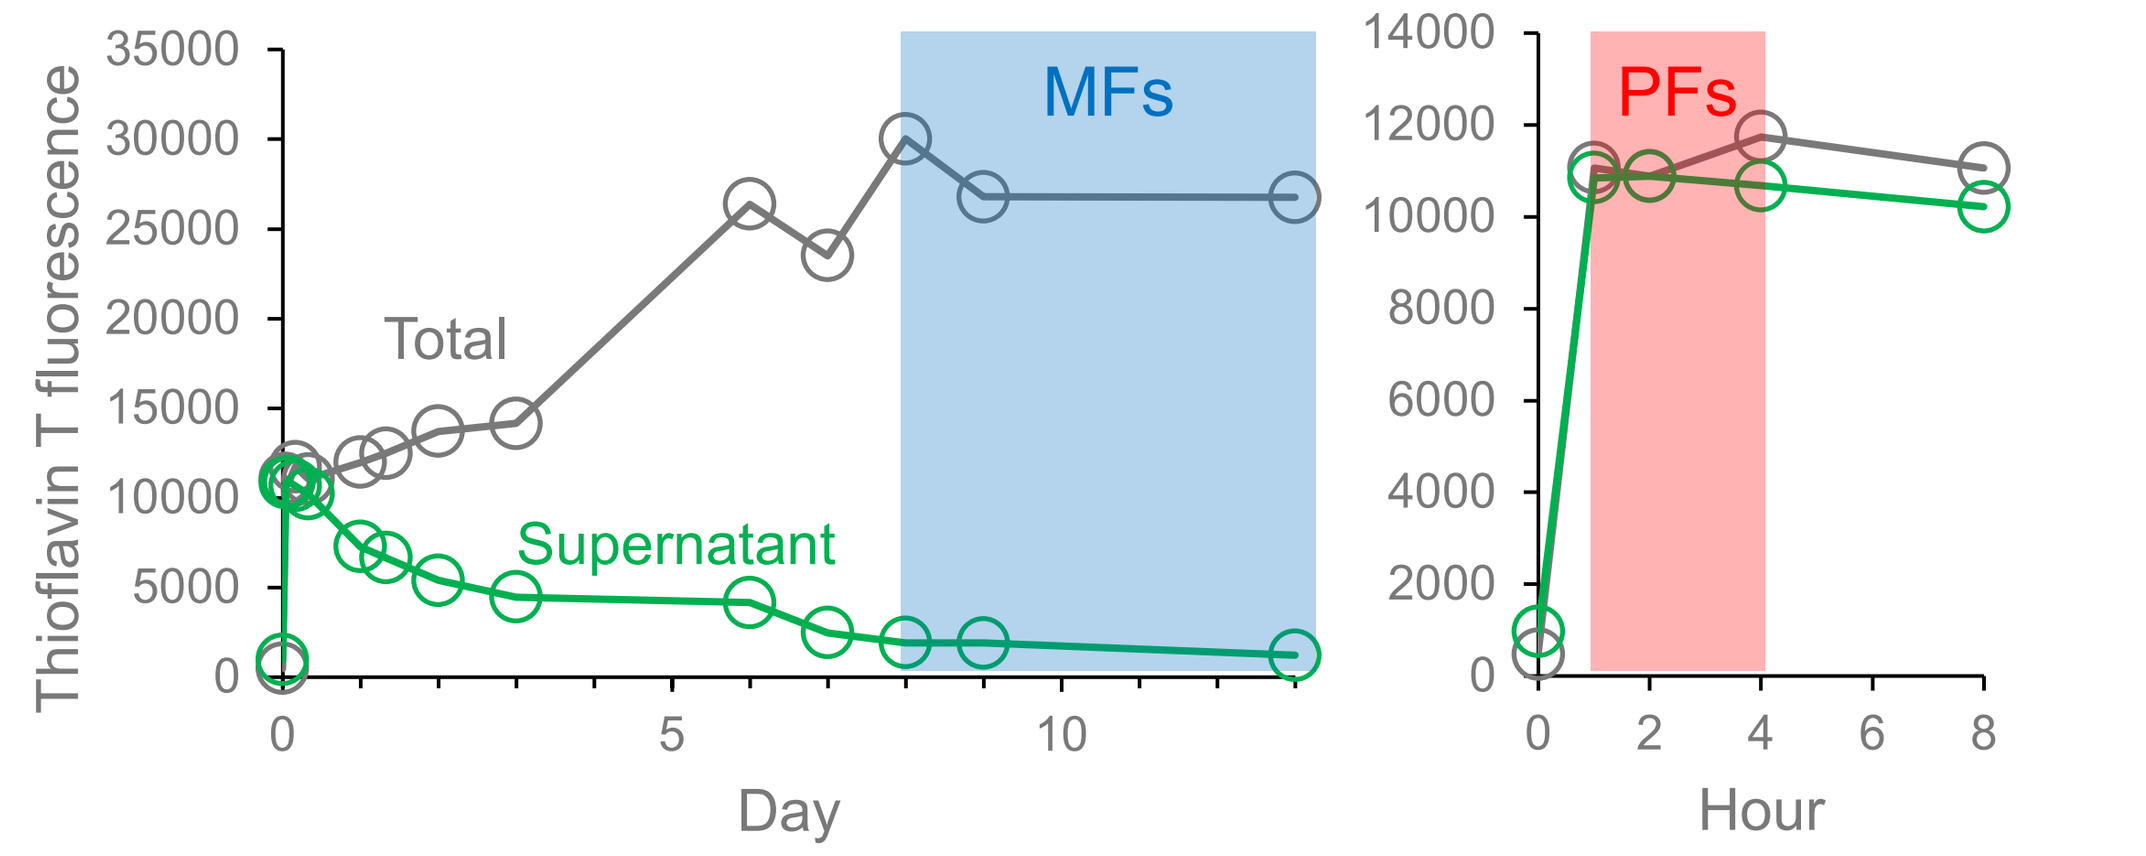

Thioflavin T fluorescence
Total
Supernatant
MFs
PFs
Day
Hour

Figure 2. Thioflavin T fluorescence time courses for soluble (supernatant) and total fractions of Aβ1-42 peptide incubation.

HS-AFM observation (Fig. 3)

1. **Structural dynamics of PFs and MFs.** PFs or MFs were deposited on the HS–AFM stage. Structural dynamics was observed in phosphate buffered saline (PBS). Young's moduli of the individual aggregates were measured by force spectrometry in PBS. The anti–Aβ antibody (Ab) was added in the chamber, followed by HS–AFM observation.
2. **Structural specificity of anti–Aβ Abs.** MFs including PFs were deposited on Ab-coated HS–AFM stage. After removal of unbound aggregates, bound aggregates were observed.

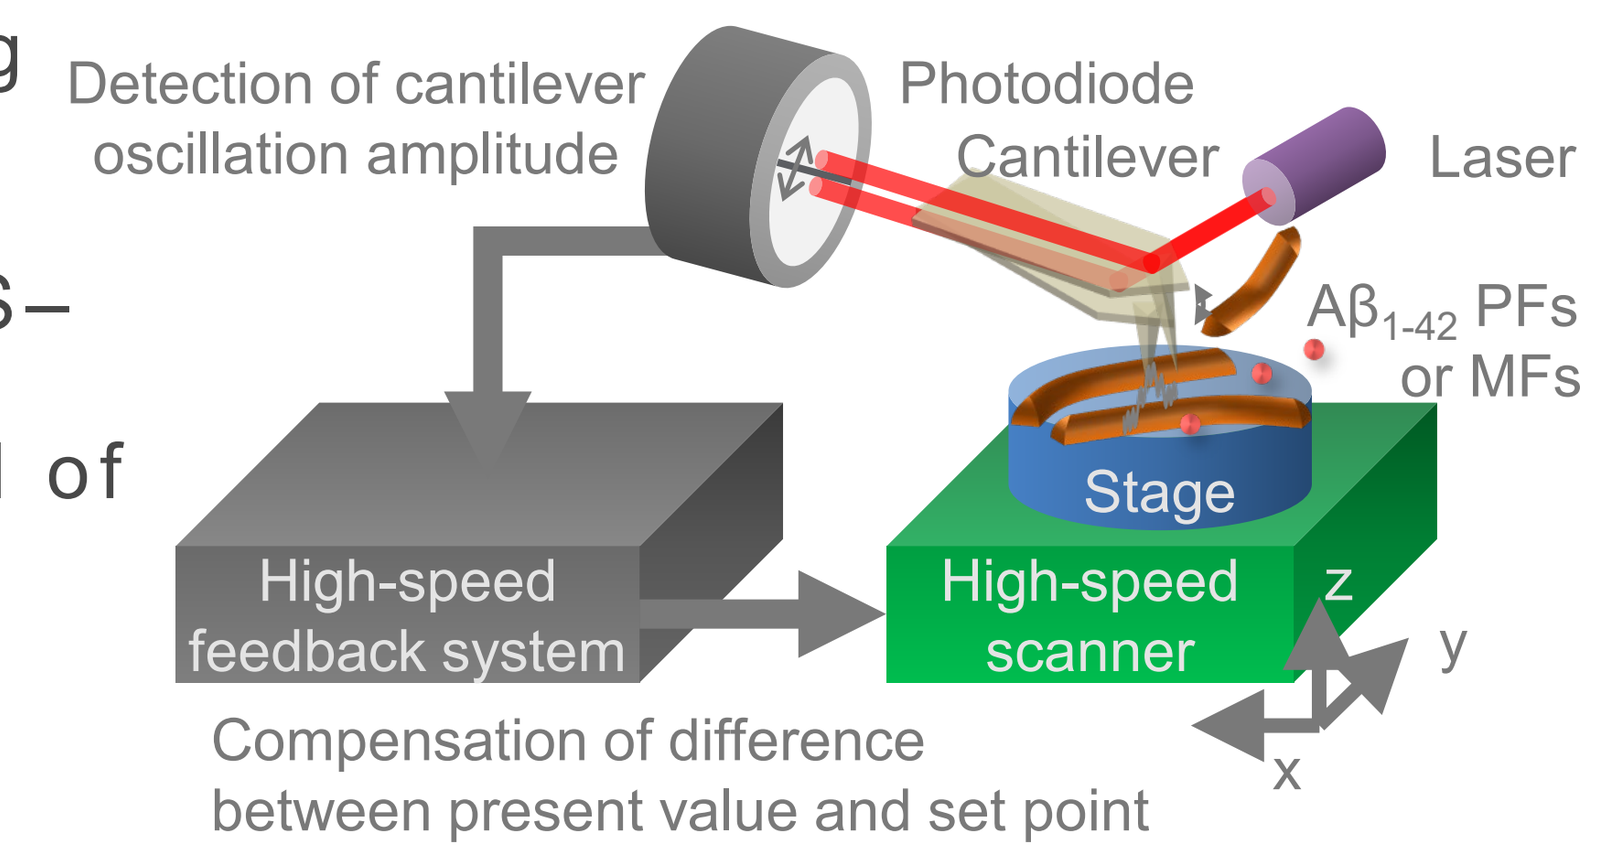

Detection of cantilever oscillation amplitude
Photodiode
Cantilever
Laser
$A\beta_{1-42}$ PFs or MFs
Stage
High-speed feedback system
High-speed scanner
Compensation of difference between present value and set point

Figure 3. HS-AFM

PFs

PF structure was composed of several globular parts in a polygonal line (Fig. 4a). The angle between each segment of a PF was maintained during diffusive motion (Fig. 4b). The length of PFs showed a single exponential distribution. The mean length depended on PF concentration (Fig. 4c).

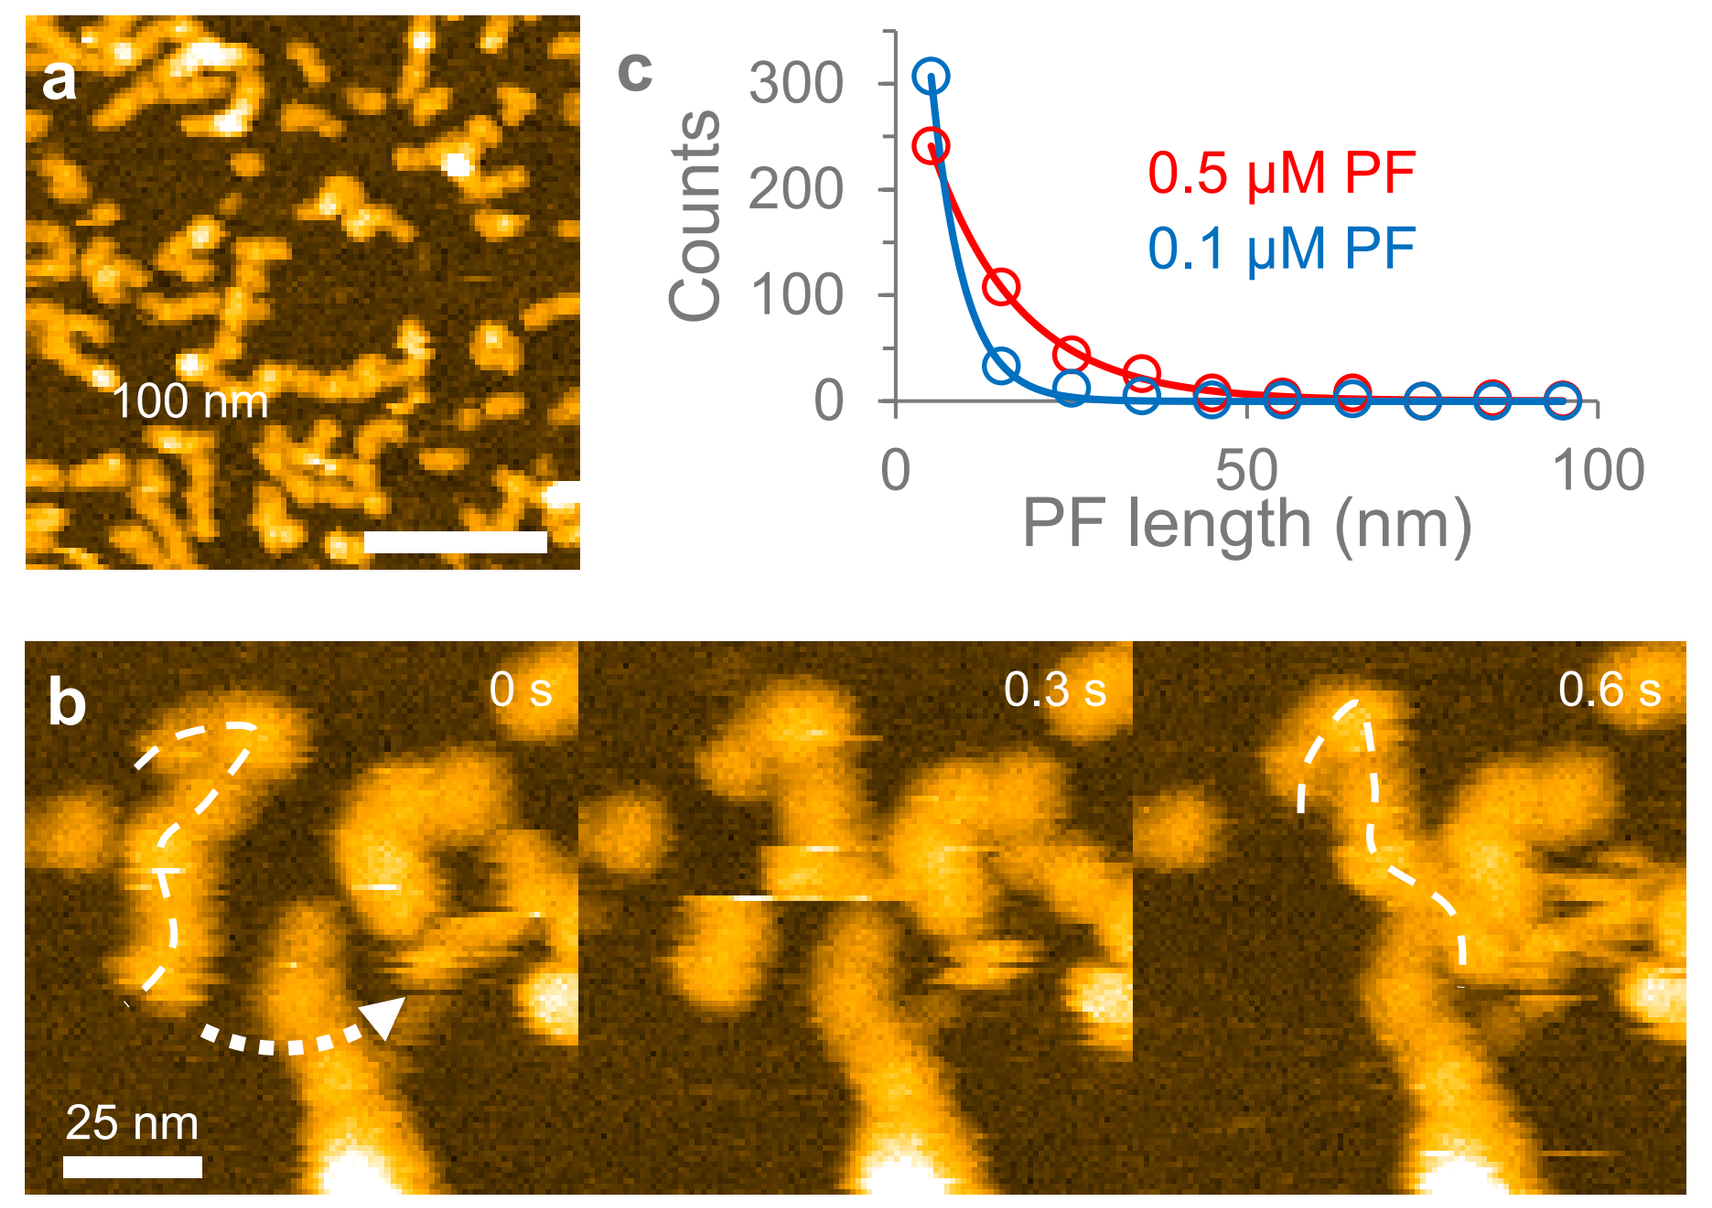

100 nm
Counts
0.5 µM PF
0.1 µM PF
PF length (nm)
0 s
0.3 s
0.6 s
25 nm

Figure 4. HS-AFM imaging of PFs.

MFs

The MF fraction contains different polymorphic fibrils with PFs (Fig. 5).

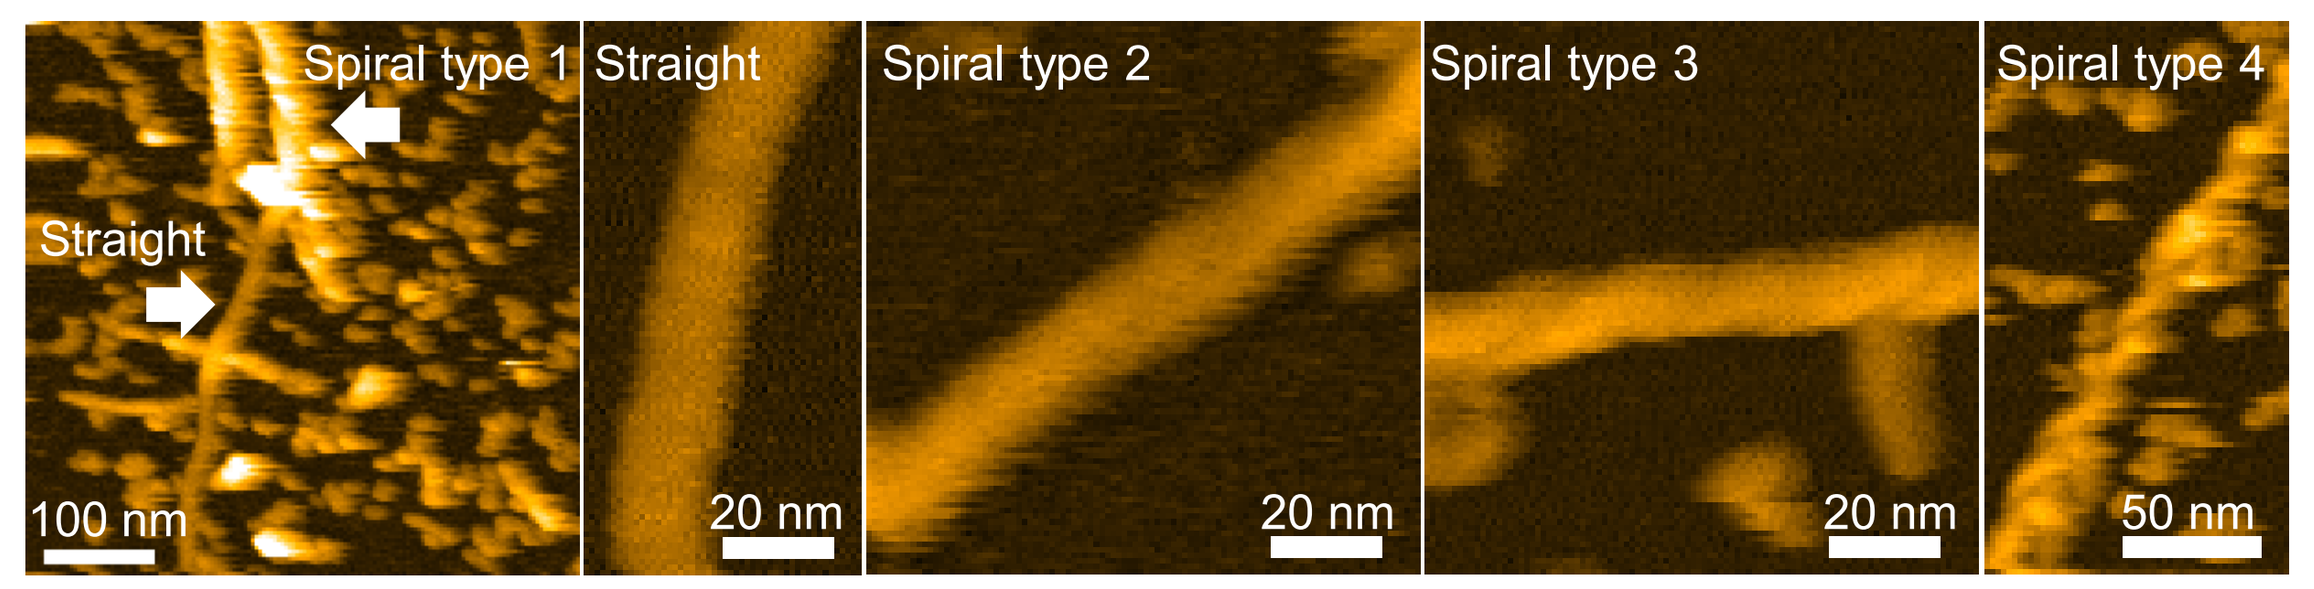

Spiral type 1
Straight
Straight
Spiral type 2
Spiral type 3
Spiral type 4
100 nm
20 nm
20 nm
20 nm
50 nm

Figure 5. HS-AFM images of MFs.

Mechanical properties of PFs and MFs

The Young's modulus of PFs was higher than that of MFs in liquid (Fig. 6).

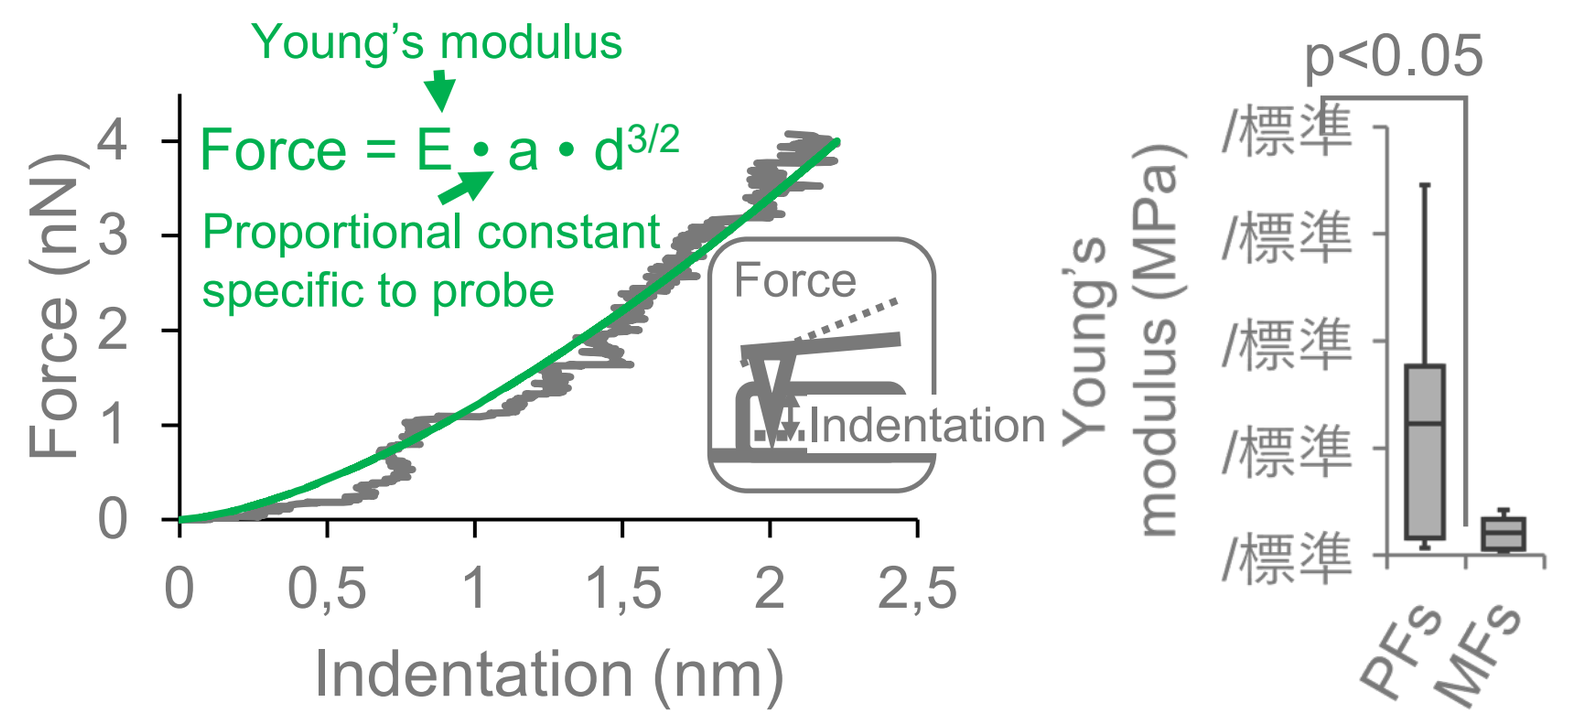

Young's modulus
Force = E • a • d^3/2
Proportional constant specific to probe
Force (nN)
Indentation (nm)
Force
Indentation
Young's modulus (MPa)
p<0.05
PFs
MFs

Figure 6. Elastic moduli of PFs and MFs (right) from force-indentation curves (left).

Antibodies vs PFs

4G8 showed poor binding to PFs (Fig. 7a), while lecanemab (BAN2401) bound(ed) to PFs rapidly and abundantly (Fig. 7b). 4G8 is a monoclonal antibody binding to the mid portion of Aβ peptide.

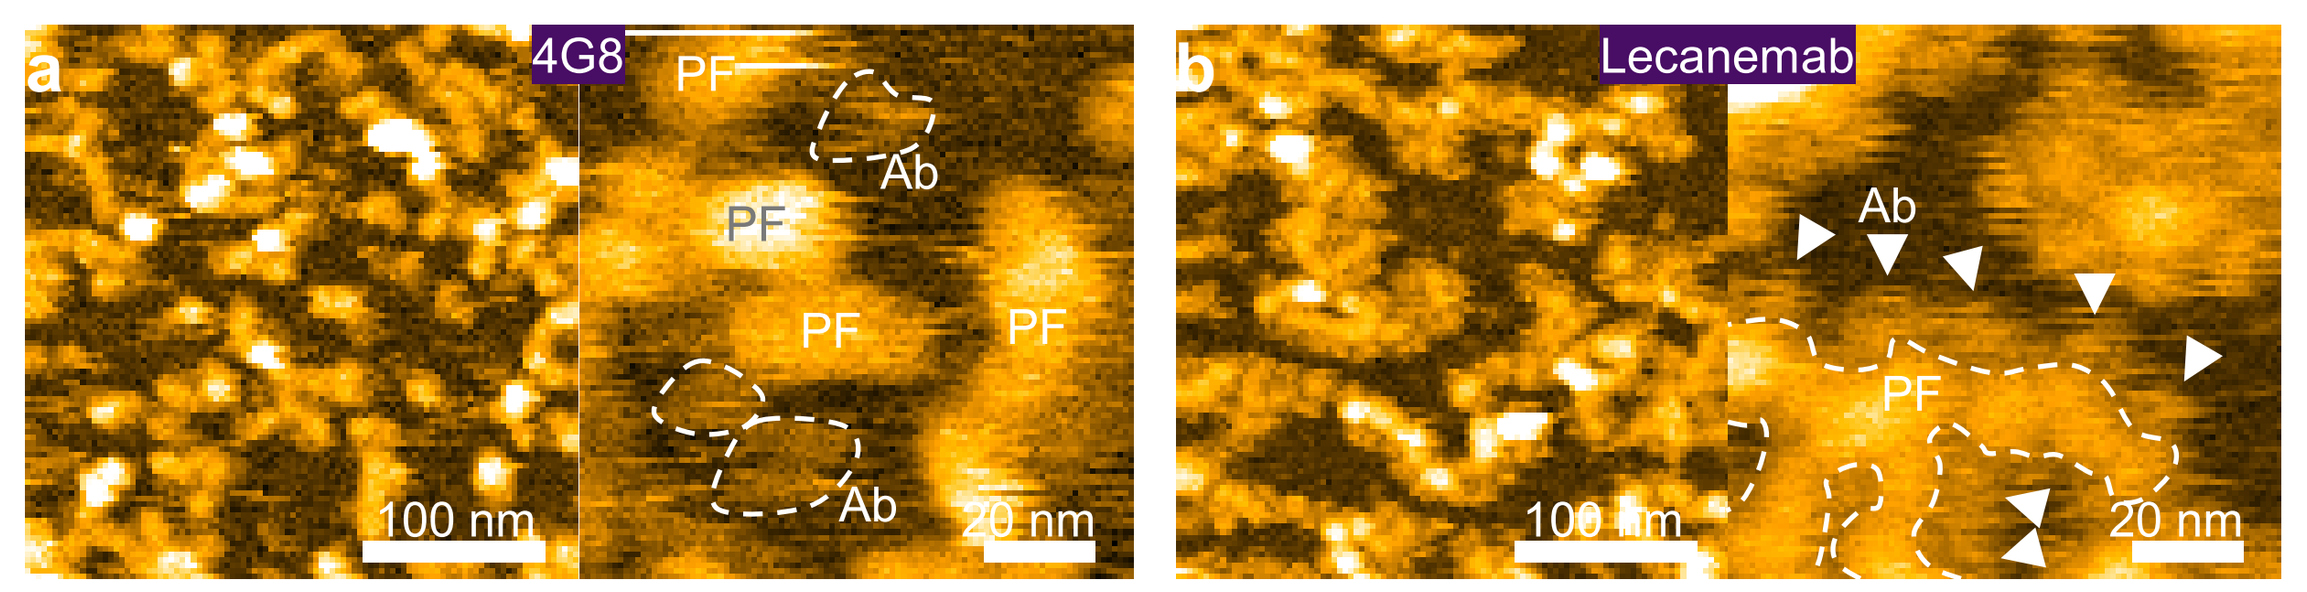

4G8
PF
Ab
Lecanemab
100 nm
20 nm

Figure 7. HS-AFM imaging of PFs with antibodies.

Antibodies vs MF fraction

4G8 bound to aggregates smaller than PFs and MFs, with minor binding to PFs in MF fraction (Fig. 8a). Lecanemab mainly bound to PFs (Fig. 8b).

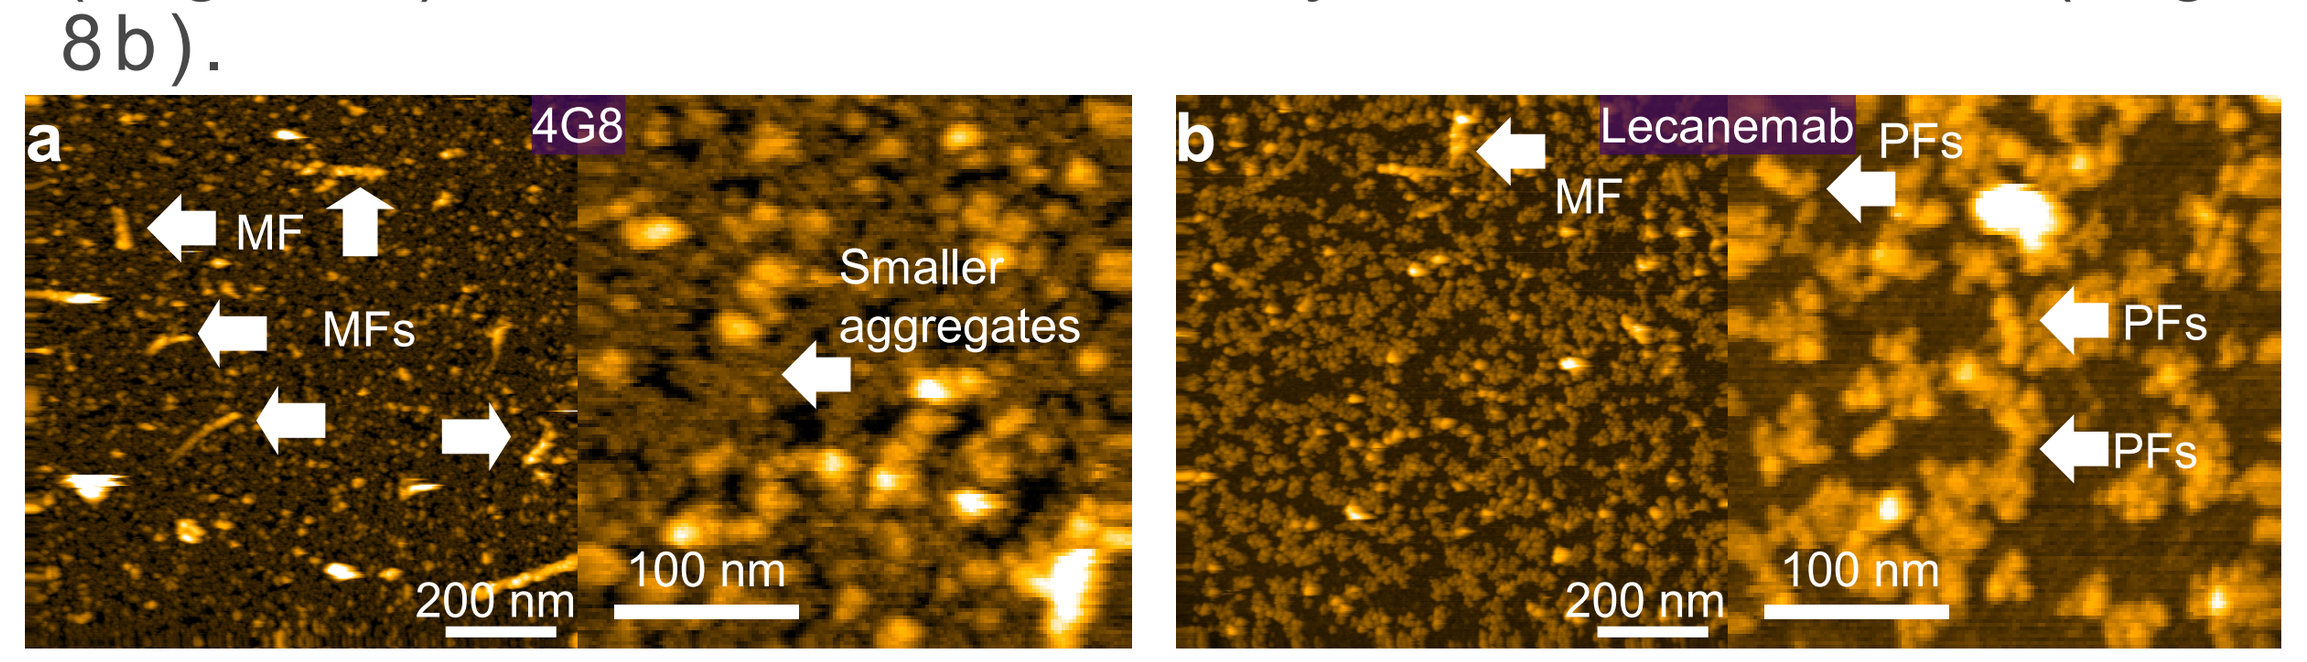

4G8
MF
MFs
Smaller aggregates
Lecanemab
PFs
200 nm
100 nm

Figure 8. HS-AFM imaging of MF fraction on antibody-coated stage.

PF length depended on its concentration, which suggests that assembly and disassembly of the globular parts in PFs are dynamic, while their segment structure is stiffer than MFs. These characteristics indicated the difference in internal structure between PFs and MFs, which supports that PFs may not be a direct precursor of MFs. Lecanemab binds to all various parts and length of PFs.

1. CJ Swanson, JL Cummings, et al. ***Alzheimer Res Therapy***. 2021
2. T. Watanabe-Nakayama, K. Ono, et al. ***Proc Natl Acad Sci USA***. 2016
3. H. Konno & T. Watanabe-Nakayama, et al. ***Proc Natl Acad Sci USA***. 2020
4. T. Watanabe-Nakayama, K. Ono ,et al. ***ACS Nano***. 2020
5. T. Yasumoto, Y. Takamura, M. Tsuji, et al. ***FASEB J***. 2019

T.W-N. tnakawata@se.kanazawa-u.ac.jp

K.O. onoken@med.showa-u.ac.jp

ALZHEIMER'S ASSOCIATION INTERNATIONAL CONFERENCE®